The Effect of Sugar Alternatives on the Drosophila Microbiome

*Kayla C. Gallant - Delaware Valley University
Dr. Cynthia Keler – Research Advisor – Delaware Valley University

Abstract

Gaining a better understanding of the gut microbiome and how it influences the overall health of an organism has become a major focus of research studies. Tests involving sugar additives are explored in this study to determine the impact of the popular substances Splenda, Stevia, and Sweet n' Low on the microbiome of *Drosophila melanogaster*, specifically *Lactobacillus species* and *Acetobacter species* levels. Populations of Drosophila were fed diets containing these substances and allowed to feed on the compounds for a week. The populations were fed hydrogen peroxide as a negative control which will kill the gut bacteria. Cornmeal which has been proven to increase *Lactobacillus sp.* populations and was used as a positive control, and fresh fruit was used as a positive control for the presence of *Acetobacter sp.* in the Drosophila microbiome. After a week, the flies were collected and stored at -70˚C until bacterial populations could be enumerated. Enumerations were performed using MRS media, selective for *Lactobacillus sp.*, and Ethanol Media, selective for *Acetobacter sp.* This was repeated so that three samples of each population were tested, the average was then taken. Flies fed fruit contained a level of bacteria one-fold less than that of the control and were the only group to contain Acetobacter sp. All three sugar alternative substances tested had reduced levels of *Lactobacillus sp.* bacteria fivefold less than that of the control sample. Upon comparison of the *Lactobacillus sp.* CFU (bacteria/mL) in each sample it was determined that all three tested sugar alternatives negatively affected *Lactobacillus sp.* bacteria in the gut microbiome of the fruit fly.

Introduction

The microbiome has become an increasingly important topic for research. Studies of the microbiome are used to gain a greater understanding of the microbiome's impact on an organism's health. Drosophila are used as model organisms for microbiome studies due to their relatively simple group of microbiota (<30 taxa), its short life cycle allows for many generations to be observed, and for easy maintainability in the lab. (Broderick and Lemaitre, 2012) Experiments involving Drosophila have been used to test the effects of diet, antibiotics, as well as other chemicals on their microbiome populations. Specifically, the bacteria Lactobacillus has been used for study since it is easily selectable in the lab for isolations (Skendzic and Keler, 2019). Manipulation of the diets in lab raised populations of Drosophila containing high levels of complex polysaccharides (cornmeal and soy flour), show an increase in the abundance of the *Lactobacillus species*. Populations that are fed diets high in sugars show a larger abundance of *Acetobacter sp*. (Douglas 2018) In the past few years, there has been an increase in the number of sugar alternatives offered to reduce the amount of sugar consumed in diet. Some of these alternatives are naturally sourced while others are artificially created and have become the subject of debate regarding their effect on health. The experiment as follows would test the question, "Do sugar alternatives/artificial sugars have an impact on the microbiome of. Drosophila".
For this experiment, a population of Drosophila maintain at DelVal were divided into seven vials. Each vial was fed fruit fly food containing one additional component of interest (i.e. sugars, polysaccharides, and sugar alternatives such as Stevia, Splenda, and Sweet'n Low (Saccharin)). This list contains two example of synthetic sweeteners (Sweet'n Low and Splenda), and one popular natural sweetener (Stevia). Diets high in sugar, the fruit diet in which apples were used, and a diet that is high in polysaccharides were also be used. The high sugar diet was used as a positive control for *Acetobacter sp.* and the polysaccharide (cornmeal) diet was a positive control for *Lactobacillus sp.*. The data gathered will be used to determine if sugar substitutes whether they be artificial or natural in a fruit fly's diet have an impact on their microbiome composition.

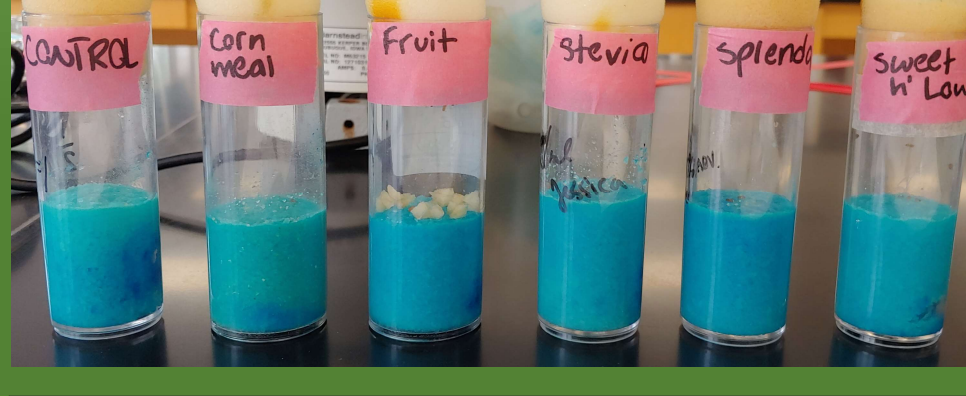
CONTROL
Corn meal
Fruit
stevia
splenda
sweet n' Low

Fruit fly tubes containing media and substance of interest (missing hydrogen peroxide – "kill" sample)

Methods and Materials

Each vial of flies was to be fed a diet composed of the fruit fly food as well as the addition of one of the substances of interest: hydrogen peroxide, cornmeal (polysaccharide), apple, Splenda, Stevia, and Sweet n' Low (Saccharin). A control group was fed only the fruit fly food, the hydrogen peroxide acted as a kill group in which no growth was expected. Flies were collected from each population after they had a week of exposure to each additive. The fly populations were stored at -70°C and stored before serial dilutions were performed. Three flies were plated from each population using serial dilution and spread plate method on two different types of plates. The first plate was MRS media to select for *Lactobacillus sp.* populations and the second plate was an ethanol media selective for the growth of *acetobacter sp*. The plates were then stored at 37˚C and 27˚ respectively. After the plates had grown out for 48 hours, the colonies were counted and the number of bacteria in the sample were enumerated.

Materials used:

- Fruit Fly Populations
- Fruit Fly Food
- Fruit Fly Vials
- MRS Media
- Ethanol Media
- Petri Dishes
- Substances of Interest (Apple, polysaccharide (cornmeal), Stevia, Splenda, and Sweet'n Low (Saccharin))

Sample Number	Additive	Selective Media Used	Run 1 CFU Bacteria/mL	Run 2 CFU Bacteria/mL	Run 3 CFU Bacteria/mL	Average CFU Bacteria/mL
1	Control	MRS	1.71×10^6	4.78×10^5	3.10×10^4	7.40×10^5
		Ethanol	0.00	0.00	0.00	0.00
2	Hydrogen Peroxide	MRS	0.00	----------	---------	0.00
		Ethanol	0.00	-----------	----------	0.00
3	Cornmeal	MRS	4.8×10^6	5.1×10^6	5.16×10^5	3.5×10^6
		Ethanol	0.00	0.00	0.00	0.00
4	Apple	MRS	8.5×10^4	2.8×10^5	7.30×10^5	3.7×10^5
		Ethanol	6.2×10^4	3.15×10^5	1.60×10^5	1.8×10^5
5	Splenda	MRS	5.1×10^4	2.31×10^5	1.98×10^5	1.6×10^5
		Ethanol	0.00	0.00	0.00	0.00
6	Stevia	MRS	8.2×10^4	4.3×10^5	6.50×10^3	1.7×10^5
		Ethanol	0.00	0.00	0.00	0.00
7	Sweet n' Low	MRS	1.34×10^4	9.4×10^4	3.20×10^5	1.4×10^5
		Ethanol	0.00	0.00	0.00	0.00

On MRS plates the number of *Lactobacillus sp* colonies were counted
On Ethanol media plates the number of *Acetobacter sp* colonies were counted

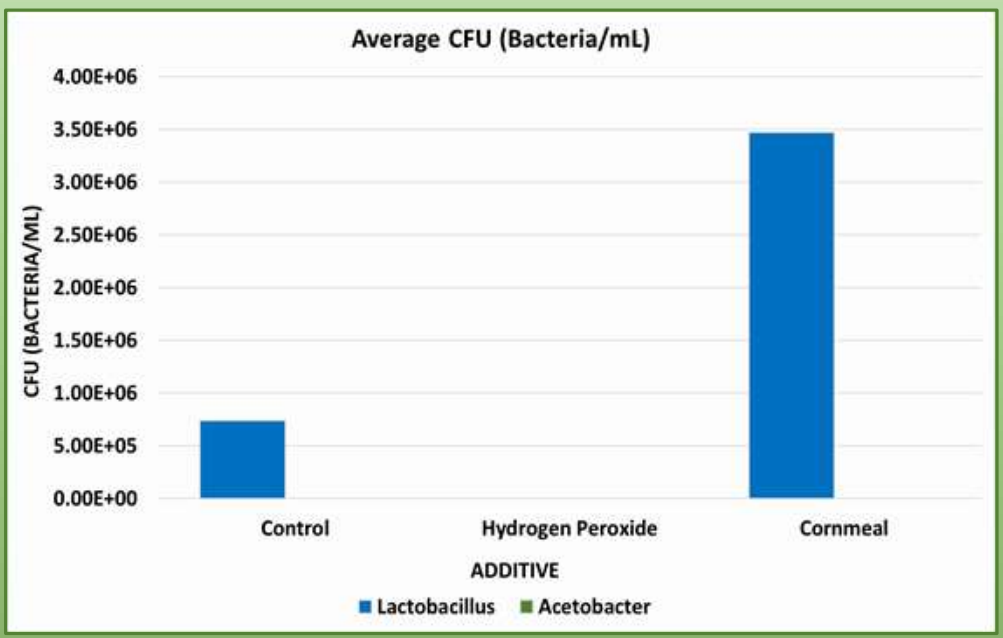
Average CFU (Bacteria/mL)
4.00E+06
3.50E+06
3.00E+06
2.50E+06
2.00E+06
1.50E+06
1.00E+06
5.00E+05
0.00E+00
CFU (BACTERIA/ML)
Control
Hydrogen Peroxide
Cornmeal
ADDITIVE
Lactobacillus
Acetobacter

Graph 1: Comparison between control, hydrogen peroxide, and cornmeal diet. To showcase the positive effect cornmeal has on *Lactobacillus sp* populations

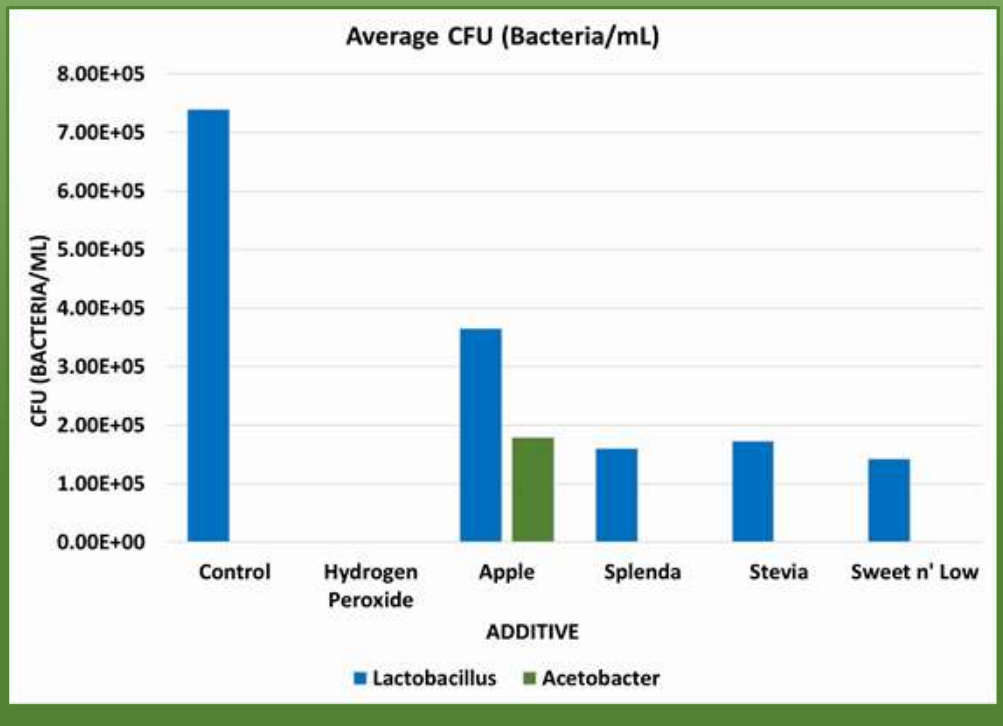
Average CFU (Bacteria/mL)
8.00E+05
7.00E+05
6.00E+05
5.00E+05
4.00E+05
3.00E+05
2.00E+05
1.00E+05
0.00E+00
CFU (BACTERIA/ML)
Control
Hydrogen Peroxide
Apple
Splenda
Stevia
Sweet n' Low
ADDITIVE
Lactobacillus
Acetobacter

Graph 2: Comparison between control and all other experimental populations

Results

Flies grown in standard fruit fly media contained an average of 7.40×10^5 CFU bacteria/mL of *Lactobacillus sp.* and 0 CFU bacteria/mL of *Acetobacter sp.*. The flies grown in fruit fly media containing apples contained 5.5×10^5 CFU of bacteria and levels of *Acetobacter sp.* and *Lactobacillus sp.* varied from that of the control. This sample contained 1.8×10^5 CFU of *Acetobacter sp.* and 3.7×10^5 of *Lactobacillus sp.*. The corn meal (polysaccharide) diet produced flies with higher levels of *Lactobacillus sp.* (3.5×10^6) than that of the control values. All three sugar alternatives tested produced levels of *Lactobacillus sp.,* lower than the control values. Flies exposed to Splenda, Stevia, and Sweet n' Low contained 1.6×10^5, 1.7×10^5, and 1.4×10^5 respectively.

Conclusion

Upon exposure to sugar alternatives such as Splenda, Stevia, and Sweet n' Low the *Lactobacillus sp.* colonies in the microbiome of the fruit fly experienced adverse effects. These effects were observed as a decreased in CFU of *Lactobacillus sp.*. To answer the proposed question of "Do sugar alternatives/artificial sugars have an impact on the microbiome of Drosophila?" the answer would be yes. Exposure these substances whether they be artificially produced, in the case of Splenda and Sweet n' Low or naturally sourced, in the case of Stevia, the overall impact on the *Lactobacillus sp.* colonies in the gut microbiome of Drosophila were negative.

Studies of the microbiome are used to gain a greater understanding of the microbiome's impact on an organism's health. As noted in an article written by Caitlin A. Brennan and Wendy S. Garrett, trends regarding the levels and health of microbiota play an important role in the prevention of diseases as well as cancer such as colorectal cancer. Gaining further knowledge about how food additives such as the sugar alternatives tested impact various microbiota and overall gut health of an organism may lead to greater understanding of how to prevent instances of colorectal cancer and other diseases.

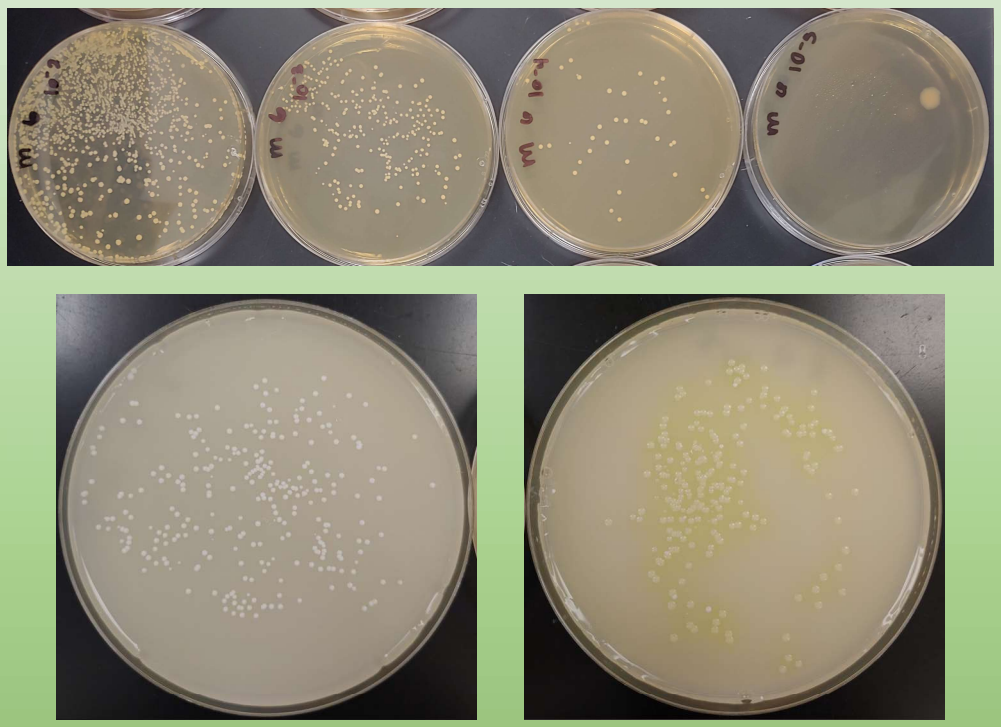

Top: Serial dilution of sample 6 (stevia) from left to right – Dilution of 10^{-2} to 10^{-5}
Bottom Left: *Lactobacillus sp.* on MRS Media
Bottom Right: *Acetobacter sp.* on Ethanol Media

Acknowledgments & Works Cited

Thank you Dr. Keler for being my mentor during this process. I appreciate your guidance and expertise greatly.

Thank you to Bristol Myer Squibb for funding this research project.

Bristol-Myers Squibb

Brennan, Caitlin A, and Wendy S Garrett. "Gut Microbiota, Inflammation, and Colorectal Cancer." *Annual review of microbiology* vol. 70 (2016): 395-411. doi:10.1146/annurev-micro-102215-095513

Broderick NA, Lemaitre B. Gut-associated microbes of Drosophila melanogaster. Gut Microbes. 2012;3(4):307-321. doi:10.4161/gmic.19896

Douglas AE. The Drosophila model for microbiome research. Lab Anim (NY). 2018;47(6):157-164. doi:10.1038/s41684-018-0065-0

Elizabeth Skendzic, Cynthia Keler; Fruit Flies & the Gut Microbiome: Redesign-Your- Bacteria Lab Exercise. The American Biology Teacher 1 January 2019; 81 (1): 47– 51. doi: https://doi.org/10.1525/abt.2019.81.1.47